Evaluation of a Single-well Cytochrome CYP3A4/5, 1A2 and 2B6 Induction Assay Using Hepatocytes Derived from HepaRG™ Cell Line

Rachel Mandell Basile[1], Gary Park[1], David Sweeny[1], Maurice Standlee[1], Susan Grepper[2], David Lau[1]

[1]Department of Drug Metabolism and Pharmacokinetics, Rigel Pharmaceuticals, South San Francisco, CA

[2]CellzDirect/Life Technologies, Durham, NC

Introduction

- Induction of drug-metabolizing enzymes can represent a major hurdle in drug development.
- Many drugs are enzyme inducers, causing time-dependent changes in pharmacokinetics of drugs metabolized by the induced pathways.
- Enzyme induction may be species-specific; therefore cryopreserved human hepatocytes are commonly used to evaluate potential for enzyme induction.
- A cell line (HepaRG™) was shown to express PXR, CAR and AhR nuclear receptors, and was shown to be inducible with known CYP inducers.

Objectives

- To evaluate and implement a simple, data-rich assay for predicting potential CYP450 induction in humans using the HepaRG™ cell line.
- To demonstrate reproducibility of the assay using CYP isozyme-specific inducers
- To compare the extent of induction, based on CYP activity and mRNA quantitation.

Methods

- Cryopreserved HepaRG™ cells, Williams Medium E (WEM), Collagen I Coated 96-well Plates, media and supplements were each obtained from Life Technologies.
- After 72 hours of exposure to inducers, hepatocytes were incubated with three probe substrates [testosterone 200 µM (3A4), bupropion 500 µM (2B6) and phenacetin 100 µM (1A2)]. Formation of 6β-hydroxy testosterone, 3-hydroxy bupropion and acetaminophen at 90 minutes were evaluated, as analyzed by LC/MS/MS
- The mRNA expression of genes of interest was determined with Panomics 2.0 QuantiGene Plex technology (Panomics/Affymetrix, Fremont, CA), following the manufacturer's protocol.
- Drug metabolizing enzymes CYP3A4, CYP2B6, CYP1A2 and the “housekeeping gene” were used in this evaluation. Other mRNA data were also collected. The mRNA levels for target genes were normalized to levels of the housekeeping gene, PPIB, based on its similar expression level compared to the P450 enzymes monitored.

Results

Figure 1. Time-linear formation of CYP2B6, CYP3A4 and CYP1A2 specific metabolites in induced HepRG™ cells

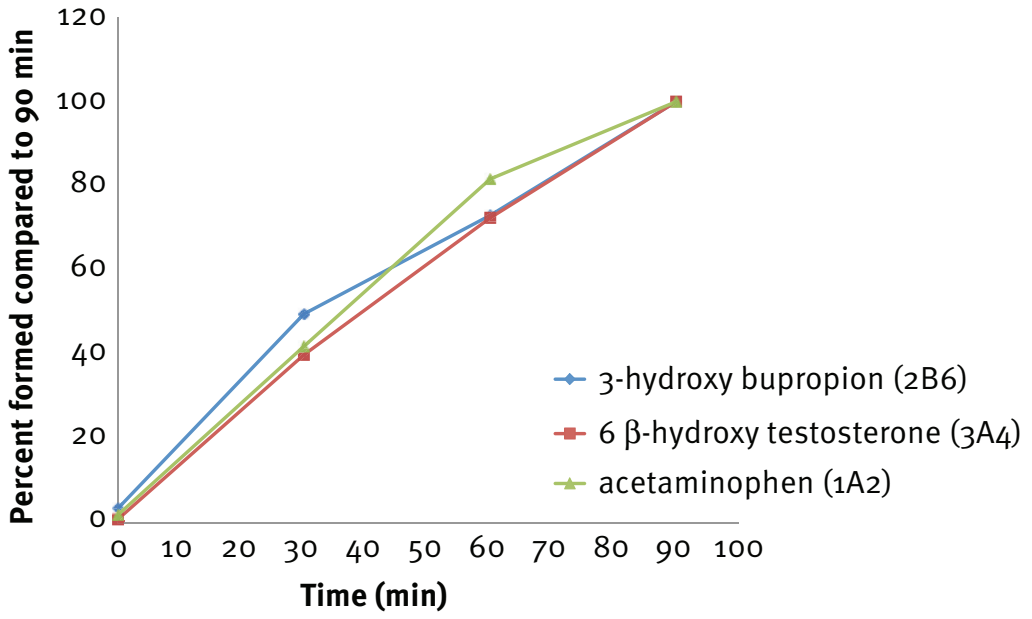
Percent formed compared to 90 min
Time (min)
0
10
20
30
40
50
60
70
80
90
100
120
3-hydroxy bupropion (2B6)
6 β-hydroxy testosterone (3A4)
acetaminophen (1A2)

Figure 2. Induced cells show an increase in metabolite formation compared to control (n=3)

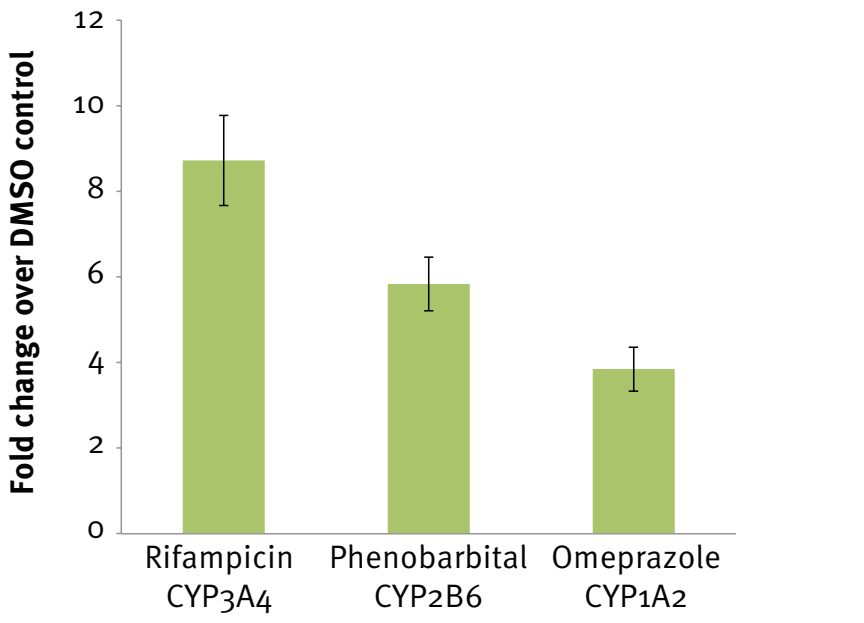
Fold change over DMSO control
0
2
4
6
8
10
12
Rifampicin CYP3A4
Phenobarbital CYP2B6
Omeprazole CYP1A2

Figure 3. Induced cells show an increase in mRNA expression compared to control (n=3)

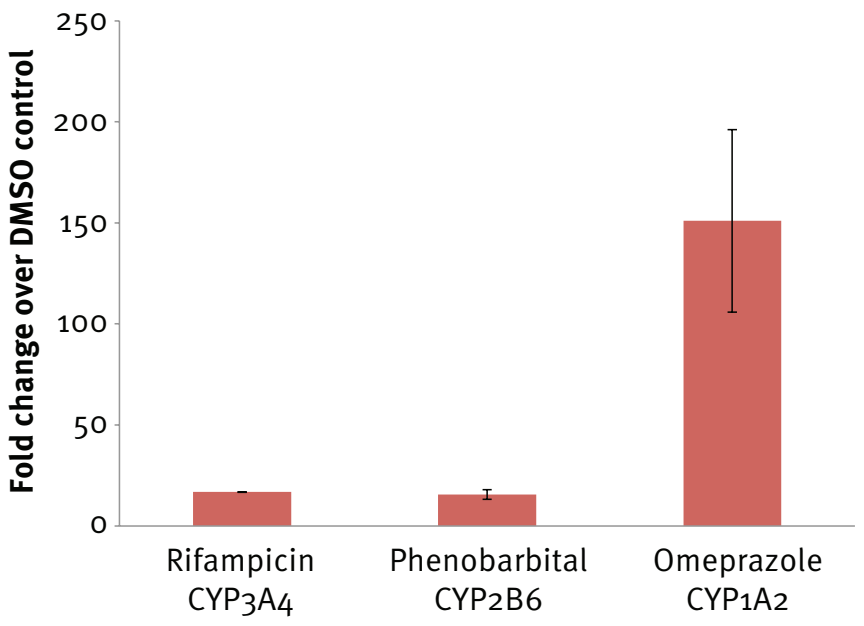
Fold change over DMSO control
0
50
100
150
200
250
Rifampicin CYP3A4
Phenobarbital CYP2B6
Omeprazole CYP1A2

Figure 4. Unexpected time-linear increase in CYP1A2 mediated acetaminophen formation by phenobarbital and rifampicin

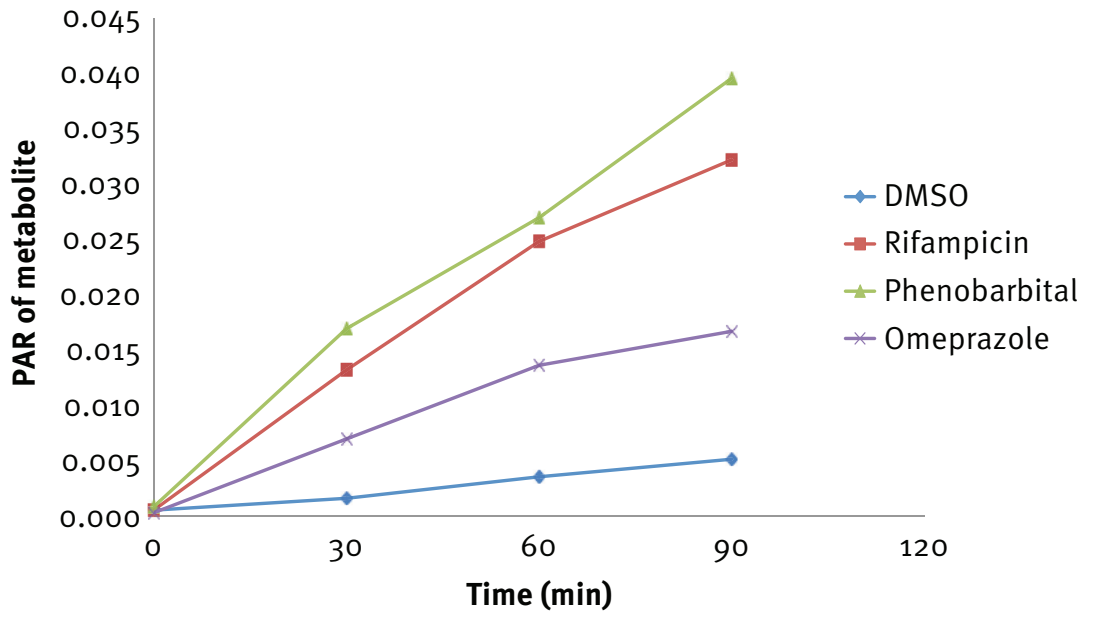
PAR of metabolite
0.000
0.005
0.010
0.015
0.020
0.025
0.030
0.035
0.040
0.045
Time (min)
0
30
60
90
120
DMSO
Rifampicin
Phenobarbital
Omeprazole

Figure 5. The increase in acetaminophen formation was not accompanied by an mRNA expression increase in the rifampicin and phenobarbital treated cells

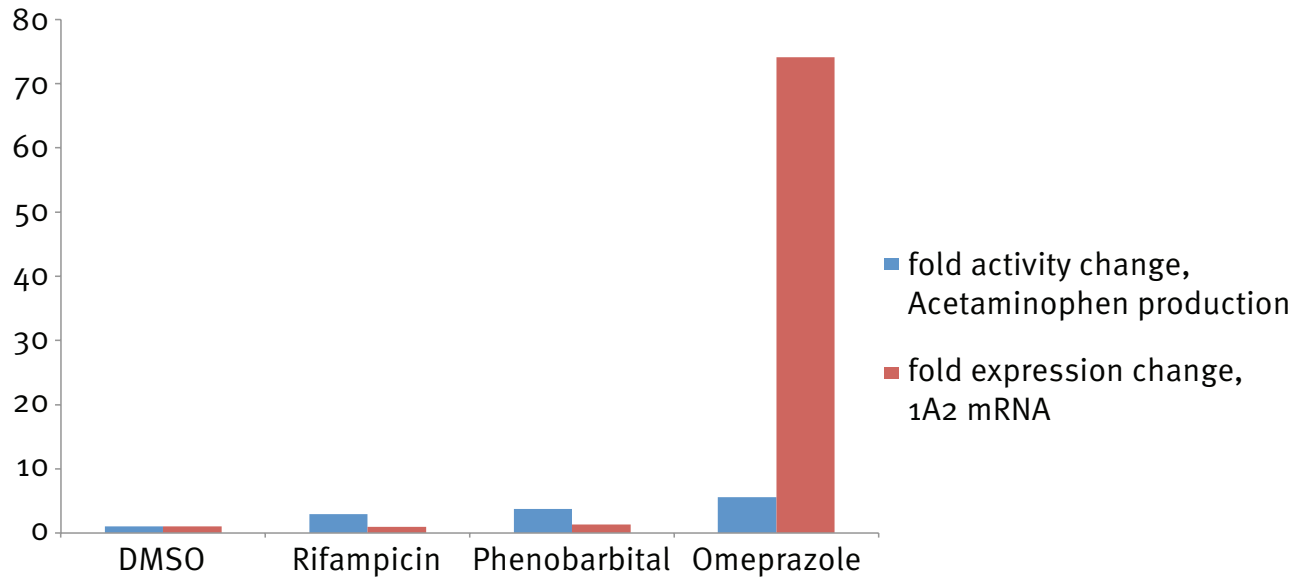
0
10
20
30
40
50
60
70
80
DMSO
Rifampicin
Phenobarbital
Omeprazole
fold activity change, Acetaminophen production
fold expression change, 1A2 mRNA

Figure 6. Effect of mixing CYP450 substrates on acetaminophen formation from phenacetin in hepatocytes treated with omeprazole, rifampicin or phenobarbital

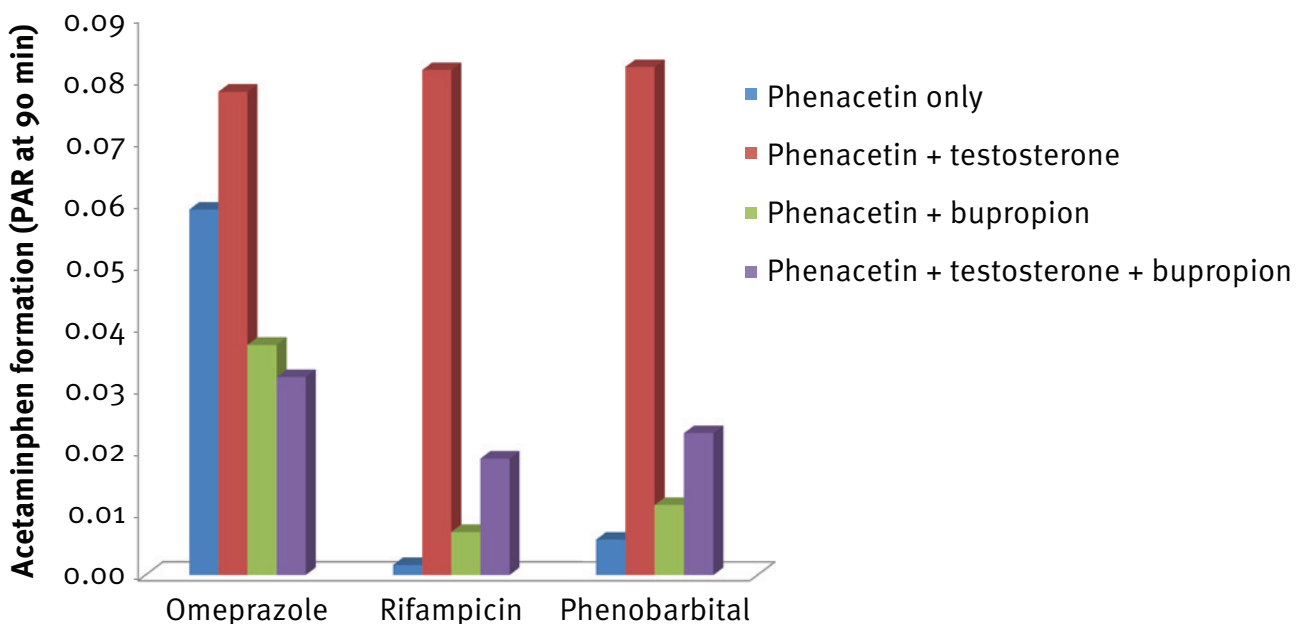
Acetaminphen formation (PAR at 90 min)
0.00
0.01
0.02
0.03
0.04
0.05
0.06
0.07
0.08
0.09
Omeprazole
Rifampicin
Phenobarbital
Phenacetin only
Phenacetin + testosterone
Phenacetin + bupropion
Phenacetin + testosterone + bupropion

Conclusions

- By using the HepaRG™ cell line, a high content assay, providing comparable results reproducibly using standards on the same plate was developed. In a single well, the induction of activity and mRNA for the three isoforms 3A4, 2B6 and 1A2 (figures 1, 2 and 3) can be evaluated.
- Correlation between the fold induction in activity and mRNA expression was obtained for positive controls for the 3A4 (rifampicin fold induction of expression/fold induction of activity=1.93), and the 2B6 isoforms (phenobarbital fold expression/fold induction=2.68).
- In the 1A2 positive control there was a larger change in expression as compared to activity for a ratio across experiments of 39.3 of expression to activity in the omeprazole induced samples (figures 2 and 3)
- Mixing the probe substrates in the activity assay, especially when phenacetin is mixed with testosterone, causes an increase in product formation of acetaminophen in rifampicin and phenobarbital induced cells (figures 4 and 6). No signal for 1A2 mRNA induction was detected in these wells (figure 5). These observations are not attributable to induction of CYP1A2 activity.

Future Work

- Evaluate cryopreserved human hepatocytes to check if the mixed substrate effect of testosterone potentiation of acetaminophen production from phenacetin O de-ethylation is present in rifampicin and phenobarbital induced cells.
- Replace testosterone with midazolam in probe substrate assay to evaluate the mixed substrate effect.
- mRNA quantitation can be used to potentially evaluate proteins that do not have defined probe substrate assays. CYP1B1, ABCB1, ABCG2 and SLC22A1 were measured for mRNA in this experiment (data not shown).
- Evaluate a similar induction assay in co-cultured animal cells that can be connected to in vivo multiple dosed studies in that same species. This could allow the prediction of time dependent changes in pharmacokinetics in animal studies.

Bibliography

Invitrogen HepaRG™ Cell User Guide, version from 2/13/2012 Affymetrix QuantiGene 2.0 Plex Assay User Manual, P/N 16659 Rev.C 2/9/2012

Cryopreserved HepaRG™ Cells: An Alternative In Vitro Screening Tool for Human Hepatic Drug Metabolism, Induction of Metabolism, & Safety Applications, Jonathan P. Jackson et al., Life Technologies, poster ISSX, 2013

An ‘all-inclusive’ 96-well cytochrome P450 induction method: Measuring enzyme activity, mRNA levels, protein levels, and cytotoxicity from one well using cryopreserved human hepatocytes, Jason S. Halladay, Susan Wong , S. Cyrus Khojasteh, Susan Grepper, Journal of Pharmacological and Toxicological Methods 66 (2012) 270–275